Tolerability and Efficacy of Crenolanib and Cytarabine/Anthracycline Chemotherapy in Older Patients (Aged 61 to 75) with Newly Diagnosed FLT3-Mutated Acute Myeloid Leukemia (AML)

Eunice S. Wang[1]; Elizabeth A. Griffiths[1]; Roland B. Walter[2]; Martin S. Tallman[3]; Aaron D. Goldberg[3]; Boo Messahel[4]; Richard M. Stone[5]

[1]Roswell Park Comprehensive Cancer Center, Buffalo, NY, United States Clinical Research Division, [2]Fred Hutchinson Cancer Research Center, Seattle, WA, [3]Memorial Sloan Kettering Cancer Center, New York, NY, [4]AROG Pharmaceuticals, Inc., Dallas, TX, [5]Dana-Farber Cancer Institute, Boston, MA.

Abstract

Background: Older AML patients often present with comorbidities and may have a compromised ability to tolerate intensive chemotherapy. These patients are more likely to have AML secondary to MDS/MPN and are considered to have a biologically distinct disease compared to their younger counterparts, with more frequent occurrence of adverse-risk cytogenetic abnormalities and mutations in genes regulating epigenetic modifications. In addition, the heterogeneity of driver mutations within a single patient contributes to limited responses to standard induction chemotherapy. While FLT3 mutations occur in this population, they are often subclonal, adding to the challenge of eradicating AML in older adult patients. Studies combining sorafenib or midostaurin with standard induction chemotherapy have shown relatively modest improvements in response rates and survival, and relapses, both early and late, remain a major concern.

There is a major unmet need for optimizing chemotherapy and TKI treatment in this medically fragile population. We here report the safety and efficacy results in newly diagnosed older patients with FLT3 mutant AML treated with crenolanib, a type I FLT3 inhibitor, in combination with intensive induction and consolidation chemotherapy (NCT02283177).

Methods: Fifteen consecutively treated patients, aged 61-75 (median age: 68), at four academic cancer centers were included in this analysis. Patients received 7+3 induction with cytarabine 100 mg/m^2 for 7 days and either daunorubicin 60 mg/m^2 or idarubicin 12 mg/m^2 for 3 days. Crenolanib 100 mg TID was administered continuously starting 24 hours after chemotherapy until 72 hours prior to the next chemotherapy cycle. Consolidation consisted of up to 4 cycles of high-dose cytarabine (HiDAC: 1 g/m^2) q12h on days 1, 3, and 5 with crenolanib starting 24 hours after the final HiDAC dose. Eligible patients proceeded to allogeneic hematopoietic stem cell transplant (HSCT). Maintenance with crenolanib at 100mg TID was started after HiDAC or 30-90 days after HSCT for up to 12 cycles.

Results: Fourteen patients completed induction chemotherapy (one patient withdrew consent at day 19). Crenolanib could be safely combined with either daunorubicin or idarubicin based induction chemotherapy. The most common adverse events (grade ≥3) were diarrhea, nausea, and febrile neutropenia. Ten of 14 patients were able to receive full doses of crenolanib during induction. The major reason for dose reduction was edema in 3 patients and GI bleeding in 1 patient. There was one treatment-related death, with 93% survival at 30 and 60 days and 87% survival at 100 days.

Complete remissions with full count recovery (CR) were achieved in 10 of 14 evaluable patients after just one cycle of induction chemotherapy. Two patients achieved a complete remission after reinduction for an overall CR rate of 86%. Of the 12 patients who achieved CR, 10 patients received HiDAC consolidation, with two patients unable to receive consolidation therapy on study. Three patients received crenolanib maintenance after multiple cycles of HiDAC consolidation. Six patients underwent HSCT and 3 received crenolanib maintenance.

As of July 2019, median OS for the intent to treat population is 20.2 months. One-year survival was 67% and 5 patients remain alive and in remission. All 5 long term survivors were ≤70 years old. All surviving patients received either multiple cycles of HiDAC or HiDAC plus transplant, and 4/5 underwent crenolanib maintenance. The patient who did not receive transplant completed 3 cycles of HiDAC consolidation and a full year of crenolanib maintenance.

Summary/Conclusion: This safety study shows that crenolanib can be combined at full doses (100mg TID) for the duration of 7+3 induction, consolidation, and maintenance in older patients with FLT3 mutant AML. Therapy was relatively well tolerated, with less than one third of patients requiring dose reductions. Long-term survival rates are encouraging in this high-risk population, but additional studies are needed to confirm the efficacy of this combination in older adults.
.

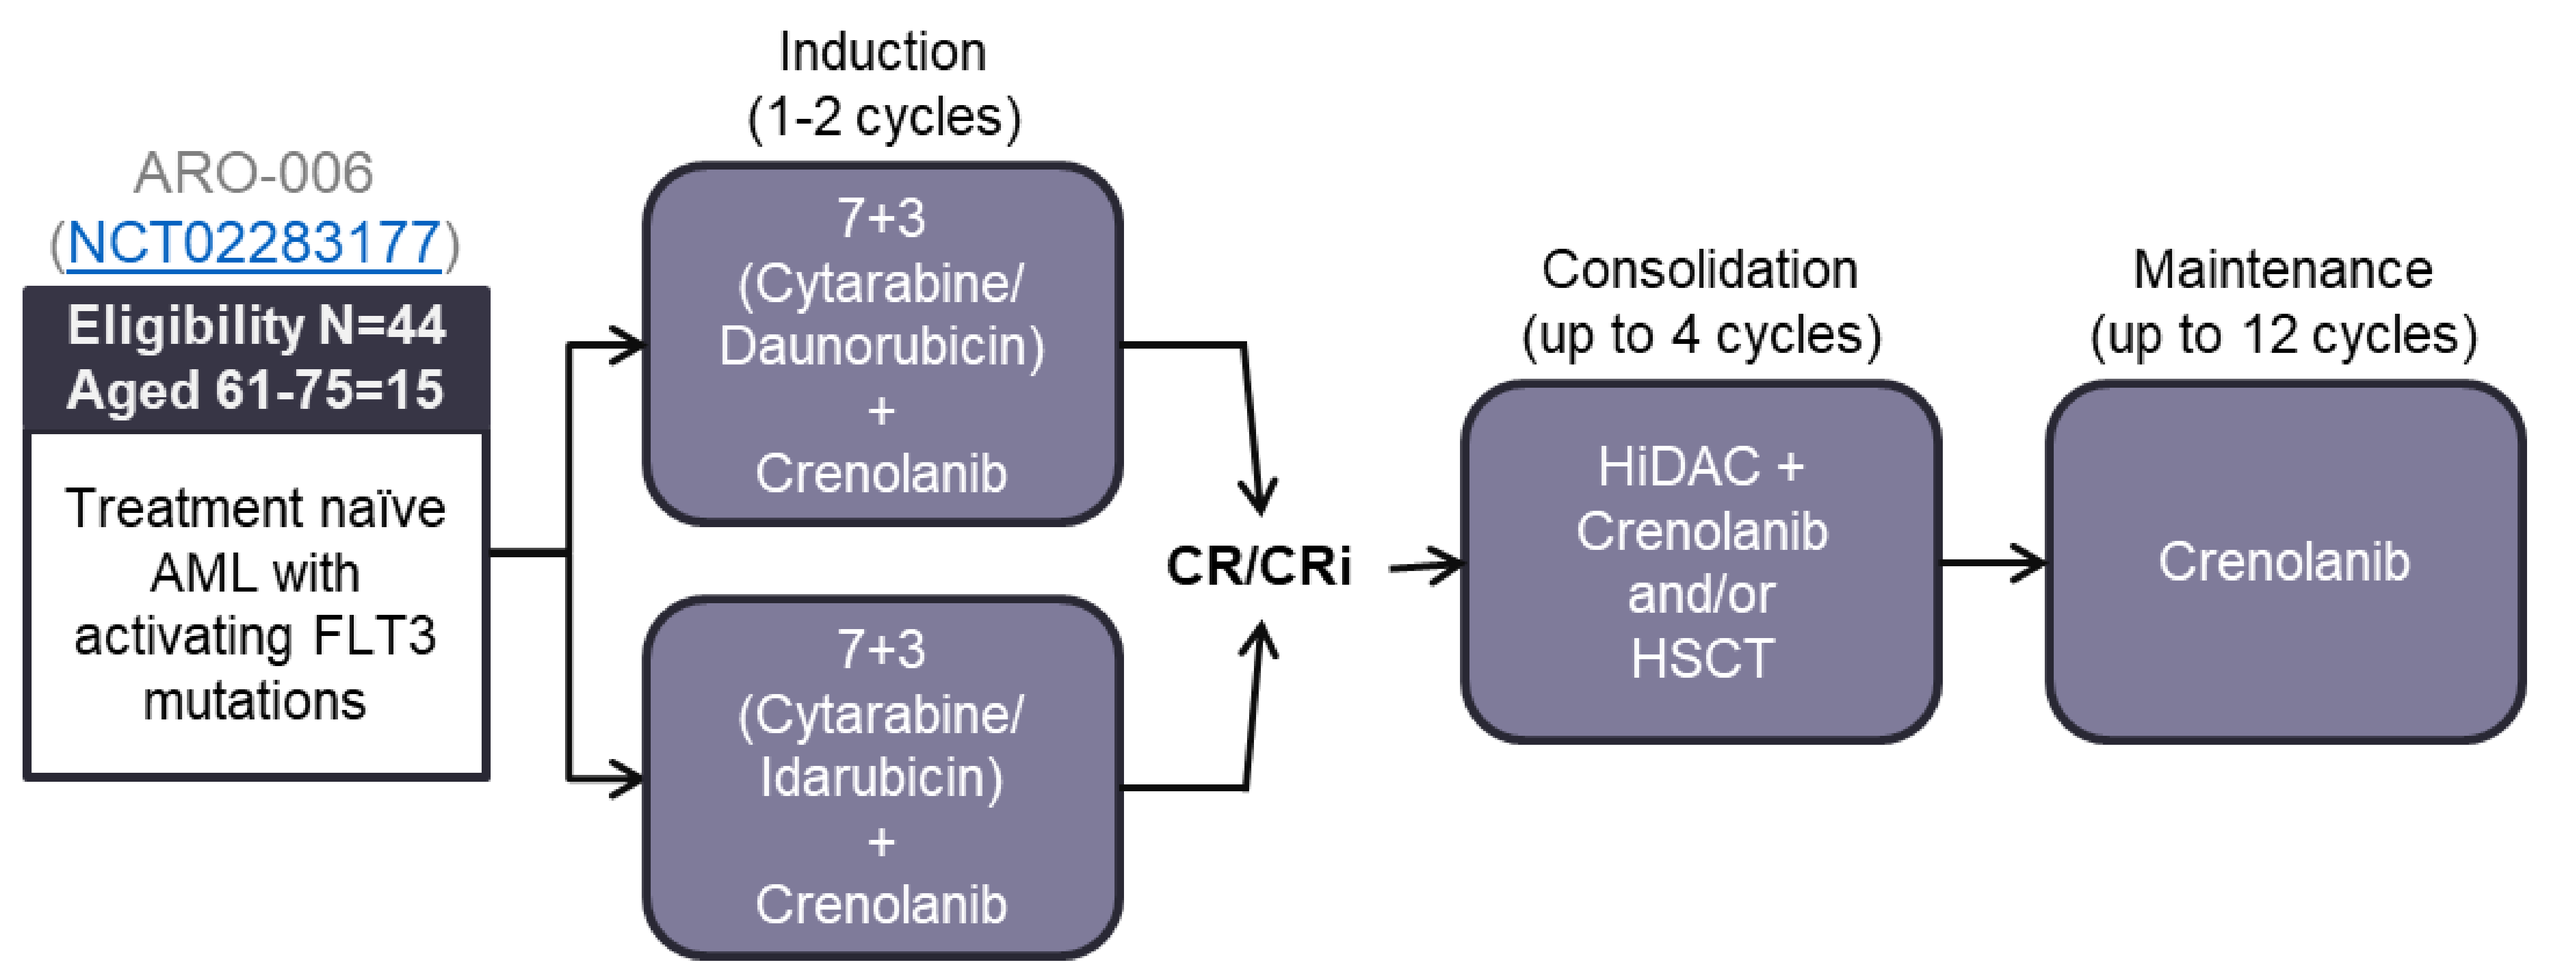
Induction
(1-2 cycles)
ARO-006
(NCT02283177)
Eligibility N=44
Aged 61-75=15
Treatment naïve AML with activating FLT3 mutations
7+3 (Cytarabine/ Daunorubicin) + Crenolanib
7+3 (Cytarabine/ Idarubicin) + Crenolanib
CR/CRi
Consolidation
(up to 4 cycles)
HiDAC + Crenolanib and/or HSCT
Maintenance
(up to 12 cycles)
Crenolanib

Background

AMLSG16-10 study of midostaurin given in combination with chemotherapy
- 78% of patients aged 60-71 years achieved a CR/CRi (n=86)
 - 48% of of patients aged 60-71 achieved a CR with full count recovery
- For patients aged 60-71, the 2-year OS was 45.6%

There remains an unmet need for therapy options in this older patient population

Sustained Full Monotherapy Crenolanib Dosing was Tolerable in Older Patients

Crenolanib was administered starting on day 9 of the chemotherapy cycle and continuing until 72 hours before the next chemotherapy cycle, allowing for sustained FLT3 inhibition, unlike other TKIs

Induction Regimen

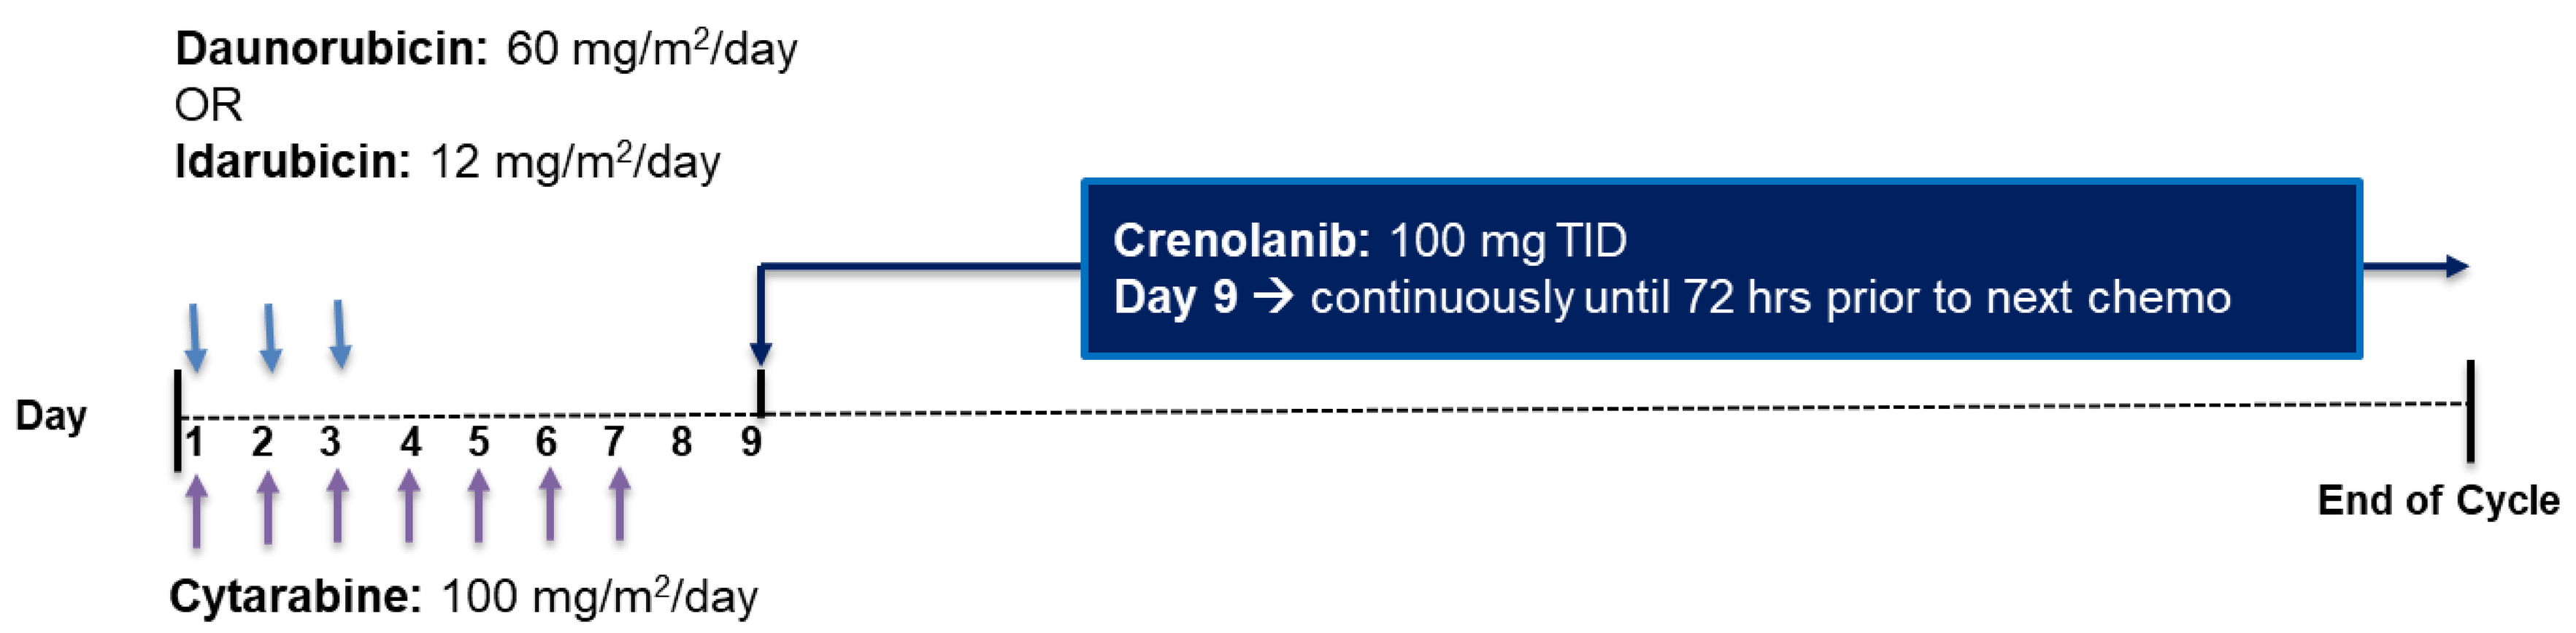
Daunorubicin: 60 mg/m2/day
OR
Idarubicin: 12 mg/m2/day
Crenolanib: 100 mg TID
Day 9 → continuously until 72 hrs prior to next chemo
Day
1 2 3 4 5 6 7 8 9
End of Cycle
Cytarabine: 100 mg/m2/day

Crenolanib tolerability with AML Induction chemotherapy				
Pt	Age/Sex	Chemotherapy	Starting Crenolanib Dose	Dose Reduction
1	68/M	Daunorubicin/cytarabine	100 TID	No
2	66/M	Idarubicin/cytarabine	100 TID	No
3	70/M	Daunorubicin/cytarabine	100 TID	No
4	61/M	Daunorubicin/cytarabine	100 TID	No
5	68/M	Daunorubicin/cytarabine	100 TID	No
6	65/M	Daunorubicin/cytarabine	100 TID	No
7	68/F	Daunorubicin/cytarabine	100 TID	No
8	61/F	Daunorubicin/cytarabine	100 TID	80 TID
9	66/F	Daunorubicin/cytarabine	100 TID	80 TID
10	69/M	Daunorubicin/cytarabine	100 TID	80 TID
11	62/F	Idarubicin/cytarabine	100 TID	No
12	74/F	Daunorubicin/cytarabine	100 TID	No
13	65/F	Daunorubicin/cytarabine	100 TID	No
14	75/M	Daunorubicin/cytarabine	100 TID	80 TID
15	74/F	Idarubicin/cytarabine	100 TID	Patient Withdrew C1D19

- 14/15 patients (93%) completed induction chemotherapy
- 11/15 patients (73%) were able to receive crenolanib at full monotherapy doses (100 mg TID) during induction
- **No patients required dose reductions for nausea, diarrhea, febrile neutropenia or delayed count recovery**
- Dose reductions were due to edema (3 patients) and GI bleeding (1 patient)

Crenolanib was Well Tolerated Following High dose Cytarabine Consolidation in Older Patients

Consolidation				
Pt	Age/Sex	Regimen (1 g/m^2 Ara-C)	Starting Crenolanib Dose	Dose Reduction
1	68/M	HiDAC + Transplant	100 TID	No
2	66/M	HiDAC	100 TID	No
3	70/M	HiDAC + Transplant	100 TID	No
4	61/M	HiDAC + Transplant	100 TID	No
5	68/M	HiDAC + Transplant	100 TID	No
6	65/M	HiDAC	100 TID	No
7	68/F	HiDAC + Transplant	100 TID	No
8	61/F	HiDAC	80 TID	No
9	66/F	HiDAC	80 TID	No
11	62/F	HiDAC + Transplant	100 TID	No

- 10/12 patients who achieved CR received consolidation
- No patients required dose reductions during consolidation

10/14 Evaluable Patients Achieved Remission with Count Recovery After 1 Cycle of Induction

71% of evaluable patients achieved a CR/CRi after one cycle of induction chemotherapy
The overall response rate to crenolanib combination therapy was 86% for evaluable patients

Induction Chemotherapy Regimen	CR after Induction 1	Overall Complete Response
Cytarabine + Daunorubicin (n=12)	8/12(67%)	10/12 (83%)
Cytarabine + Idarubicin (n=3)*	2/2 (100%)	2/2 (100%)
Total Evaluable (n=14)	10/14 (71%)	12/14 (86%)

*One patient was not evaluable and withdrew from the study before completing induction

Patients Receiving Crenolanib Combination Therapy Achieved Full Count Recover while on Crenolanib

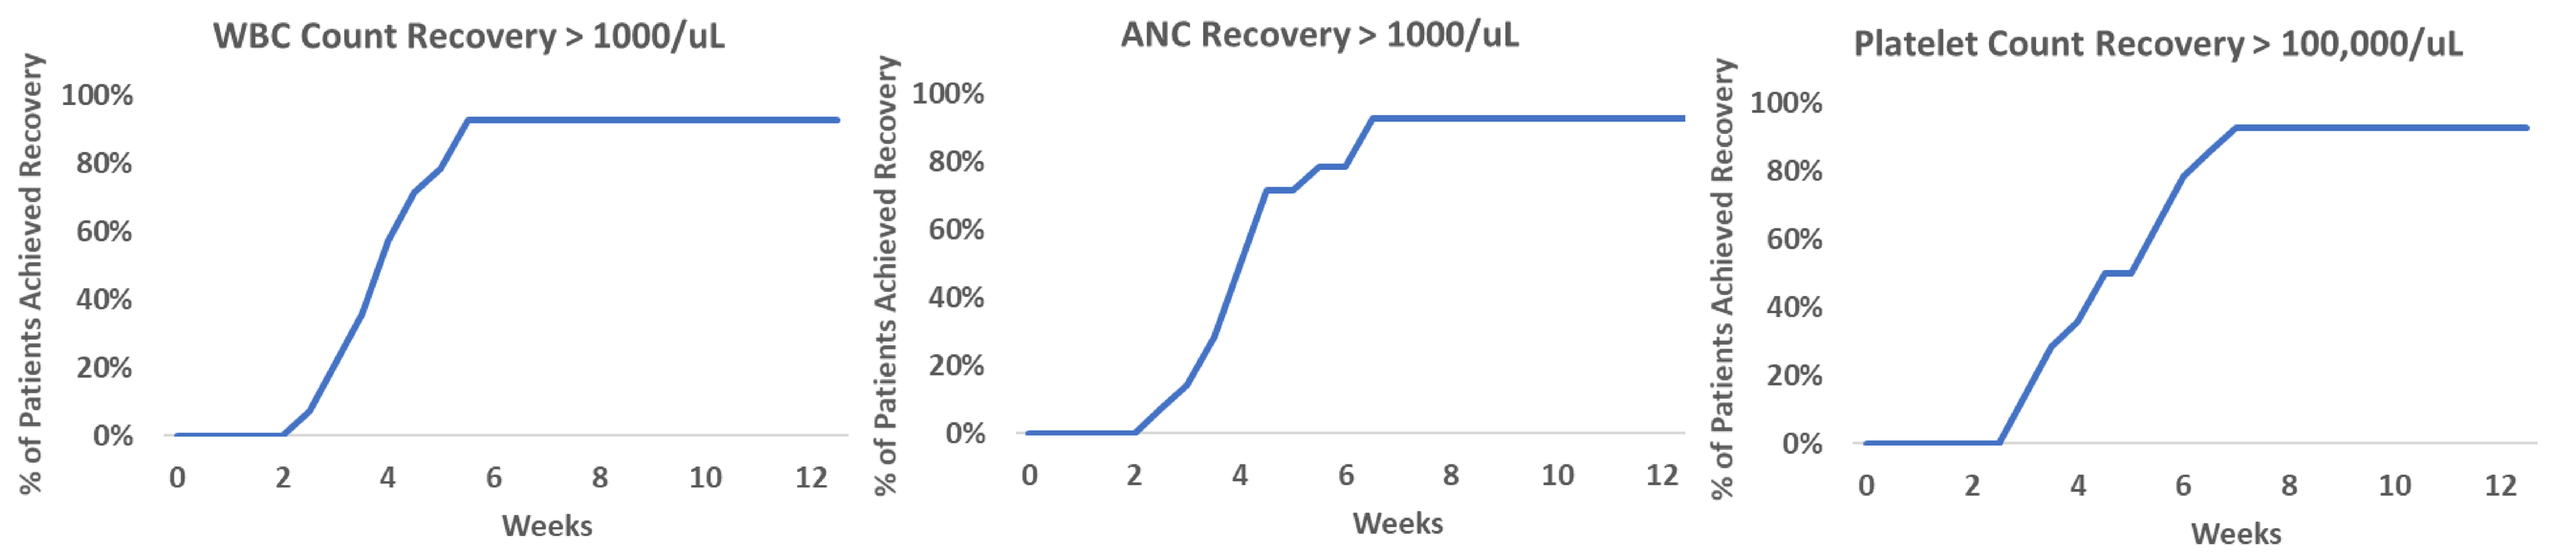
WBC Count Recovery > 1000/uL
ANC Recovery > 1000/uL
Platelet Count Recovery > 100,000/uL
% of Patients Achieved Recovery
100%
80%
60%
40%
20%
0%
0 2 4 6 8 10 12
Weeks

n=14	WBC Recovery >1,000/µL	Absolute Neutrophil Count Recovery > 500/µL	Absolute Neutrophil Count Recovery >100/µ	Platelet Recovery >100,000/µL
Median (days)	26	25	27	31

5 Patients Aged 61-70 (42%) Remain Alive and in Remission at Median 30 Months of Follow-up

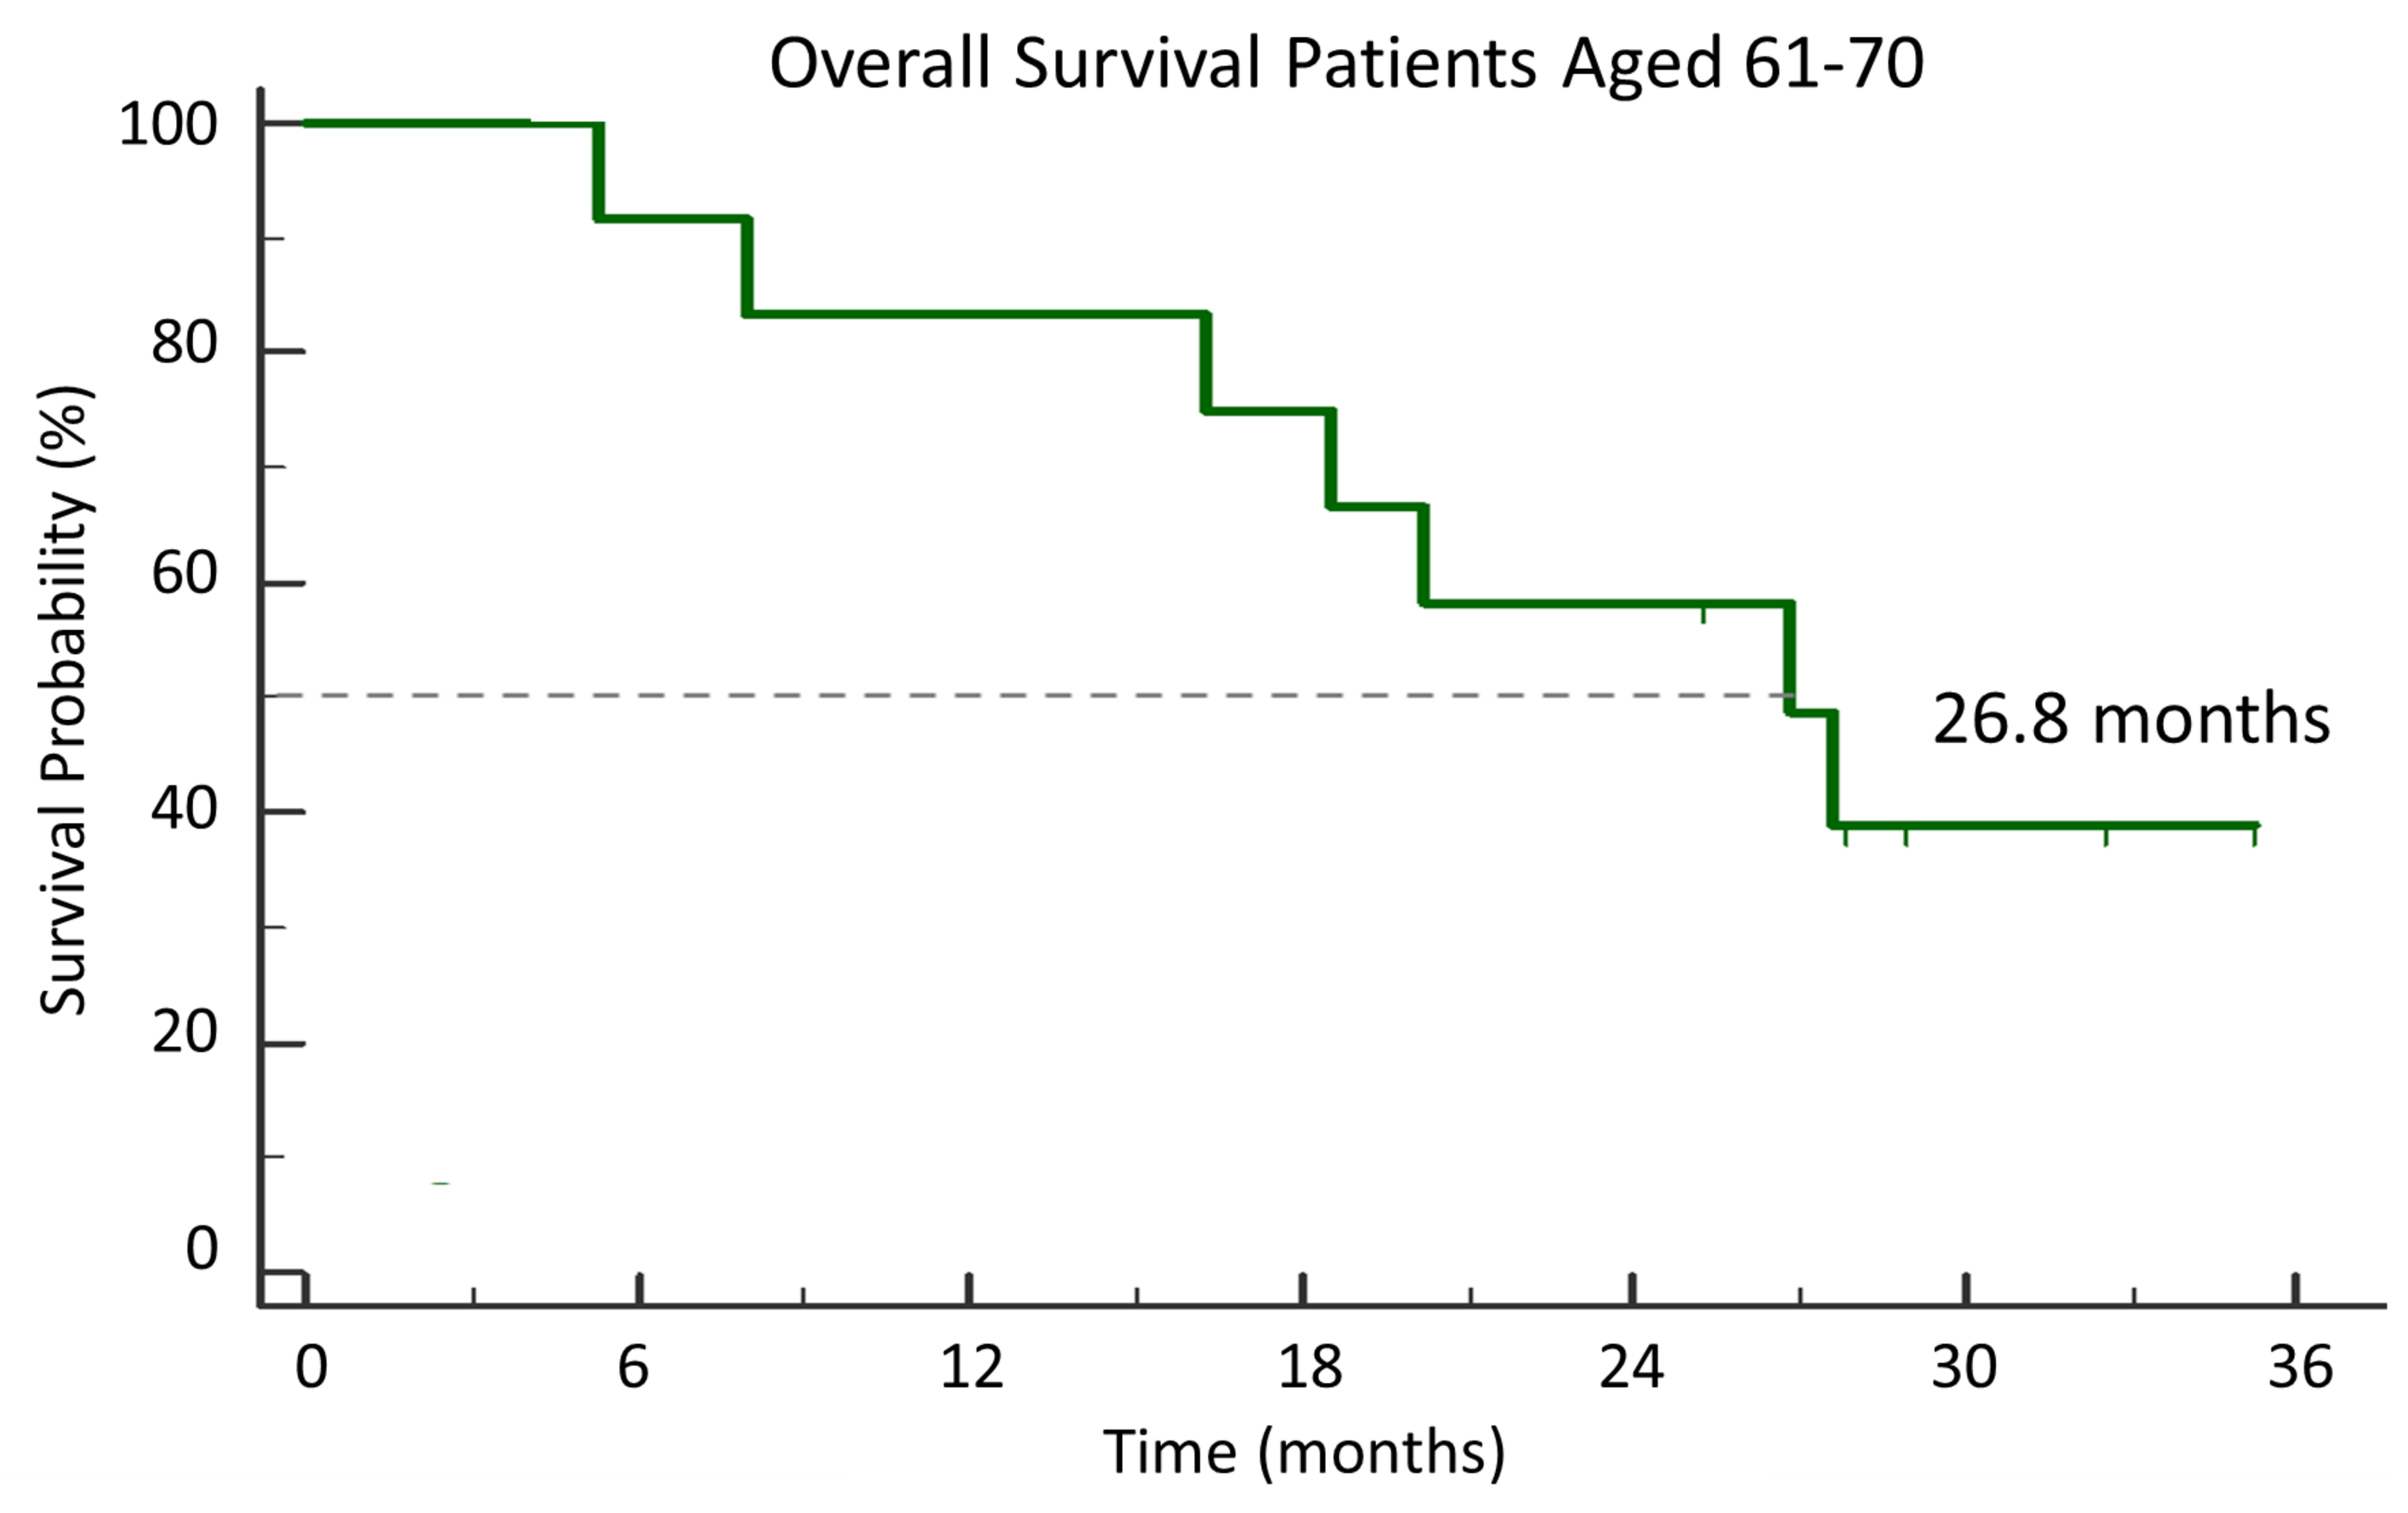
Overall Survival Patients Aged 61-70
Survival Probability (%)
100
80
60
40
20
0
0 6 12 18 24 30 36
Time (months)
26.8 months

Median OS for patients aged 61-70 (n=12) was 26.8 months

Conclusions

- These results suggest that crenolanib is safe and tolerable given sequentially with standard induction chemotherapy in older patients aged 61-75 years
- Crenolanib combination therapy provided promising overall survival rates, with low rates of early mortality

References:
Schlenk R-F Midostaurin added to chemotherapy and continued single agent maintenance therapy in acute myeloid leukemia with FLT3-ITD. *Blood 2019*